Efficacy and Safety of Telitacicept in Patients with Generalized Myasthenia Gravis: Results of a Phase 3 Study

Jian Yin,[1] Wenxiang Wang,[2] Lin Li,[2] Yan Sun,[2] Qing Zuraw,[3] Jianmin Fang[2]

[1]Department of Neurology, Beijing Hospital, Beijing, 100730, China; [2]RemeGen Co., Ltd, Yantai, Shandong 264006, China; [3]RemeGen Biosciences, Inc., San Francisco, CA, USA

Introduction

- Telitacicept is a novel recombinant fusion protein constructed with the extracellular domain of TACI and the Fc domain of human immunoglobulin (Ig) G1 (**Figure 1**).[1]
- Telitacicept inhibits B-cell activity by neutralizing B lymphocyte stimulator (BLyS) and a proliferation-inducing ligand (APRIL) (**Figure 1**).
- In generalized myasthenia gravis (gMG), B-cell produce autoantibodies targeting the acetylcholine receptor (AChR) and muscle-specific tyrosine kinase (MuSK), disrupting neuromuscular transmission, leading to muscle weakness.[1]
- Telitacicept has shown efficacy and tolerability in a phase 2 study of patients with gMG.[1]

	Binding activities		
	BAFF-R	BCMA	TACI
BLyS	++	++	+++
APRIL		++	++

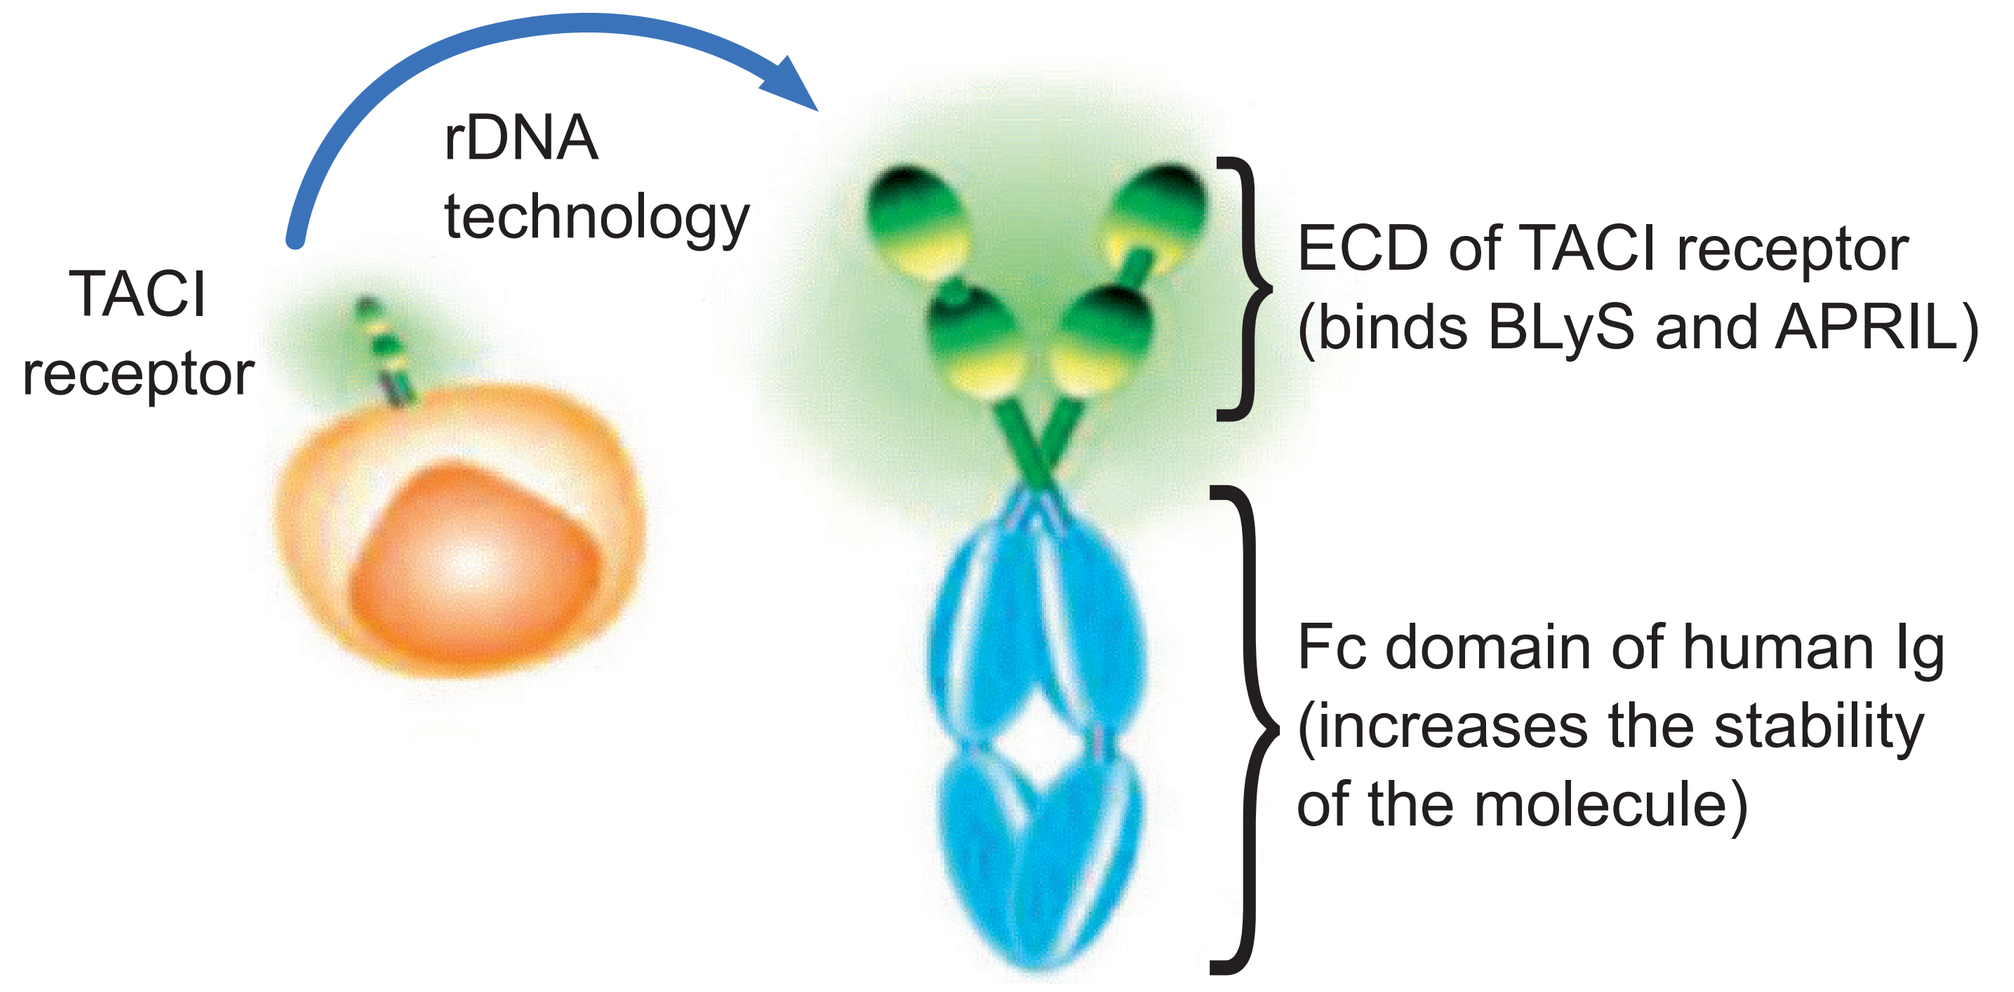
TACI receptor
rDNA technology
ECD of TACI receptor (binds BLyS and APRIL)
Fc domain of human Ig (increases the stability of the molecule)

Figure 1. Telitacicept mechanism of action. APRIL=a proliferation-inducing ligand; **BAFF-R**=B-cell activating factor receptor; **BCMA**=B-cell maturation antigen; **BLyS**=B lymphocyte stimulator; **ECD**=extracellular domain; **Ig**=immunoglobulin; **rDNA**=recombinant DNA; **TACI**=transmembrane activator and calcium modulator and cyclophilin ligand interactor.

Objective

- **To assess efficacy and safety of telitacicept in a Chinese, phase 3, multicenter, randomized, placebo-controlled study in patients with gMG.**

Methods

- **Study design:** During a 24-week double-blind period, patients were randomized 1:1 to weekly subcutaneous telitacicept (240 mg) or placebo. Completers will enter a 24-week, open-label period (weekly telitacicept [240 mg]) (**Figure 2**). We report data from the 24-week double-blind period.

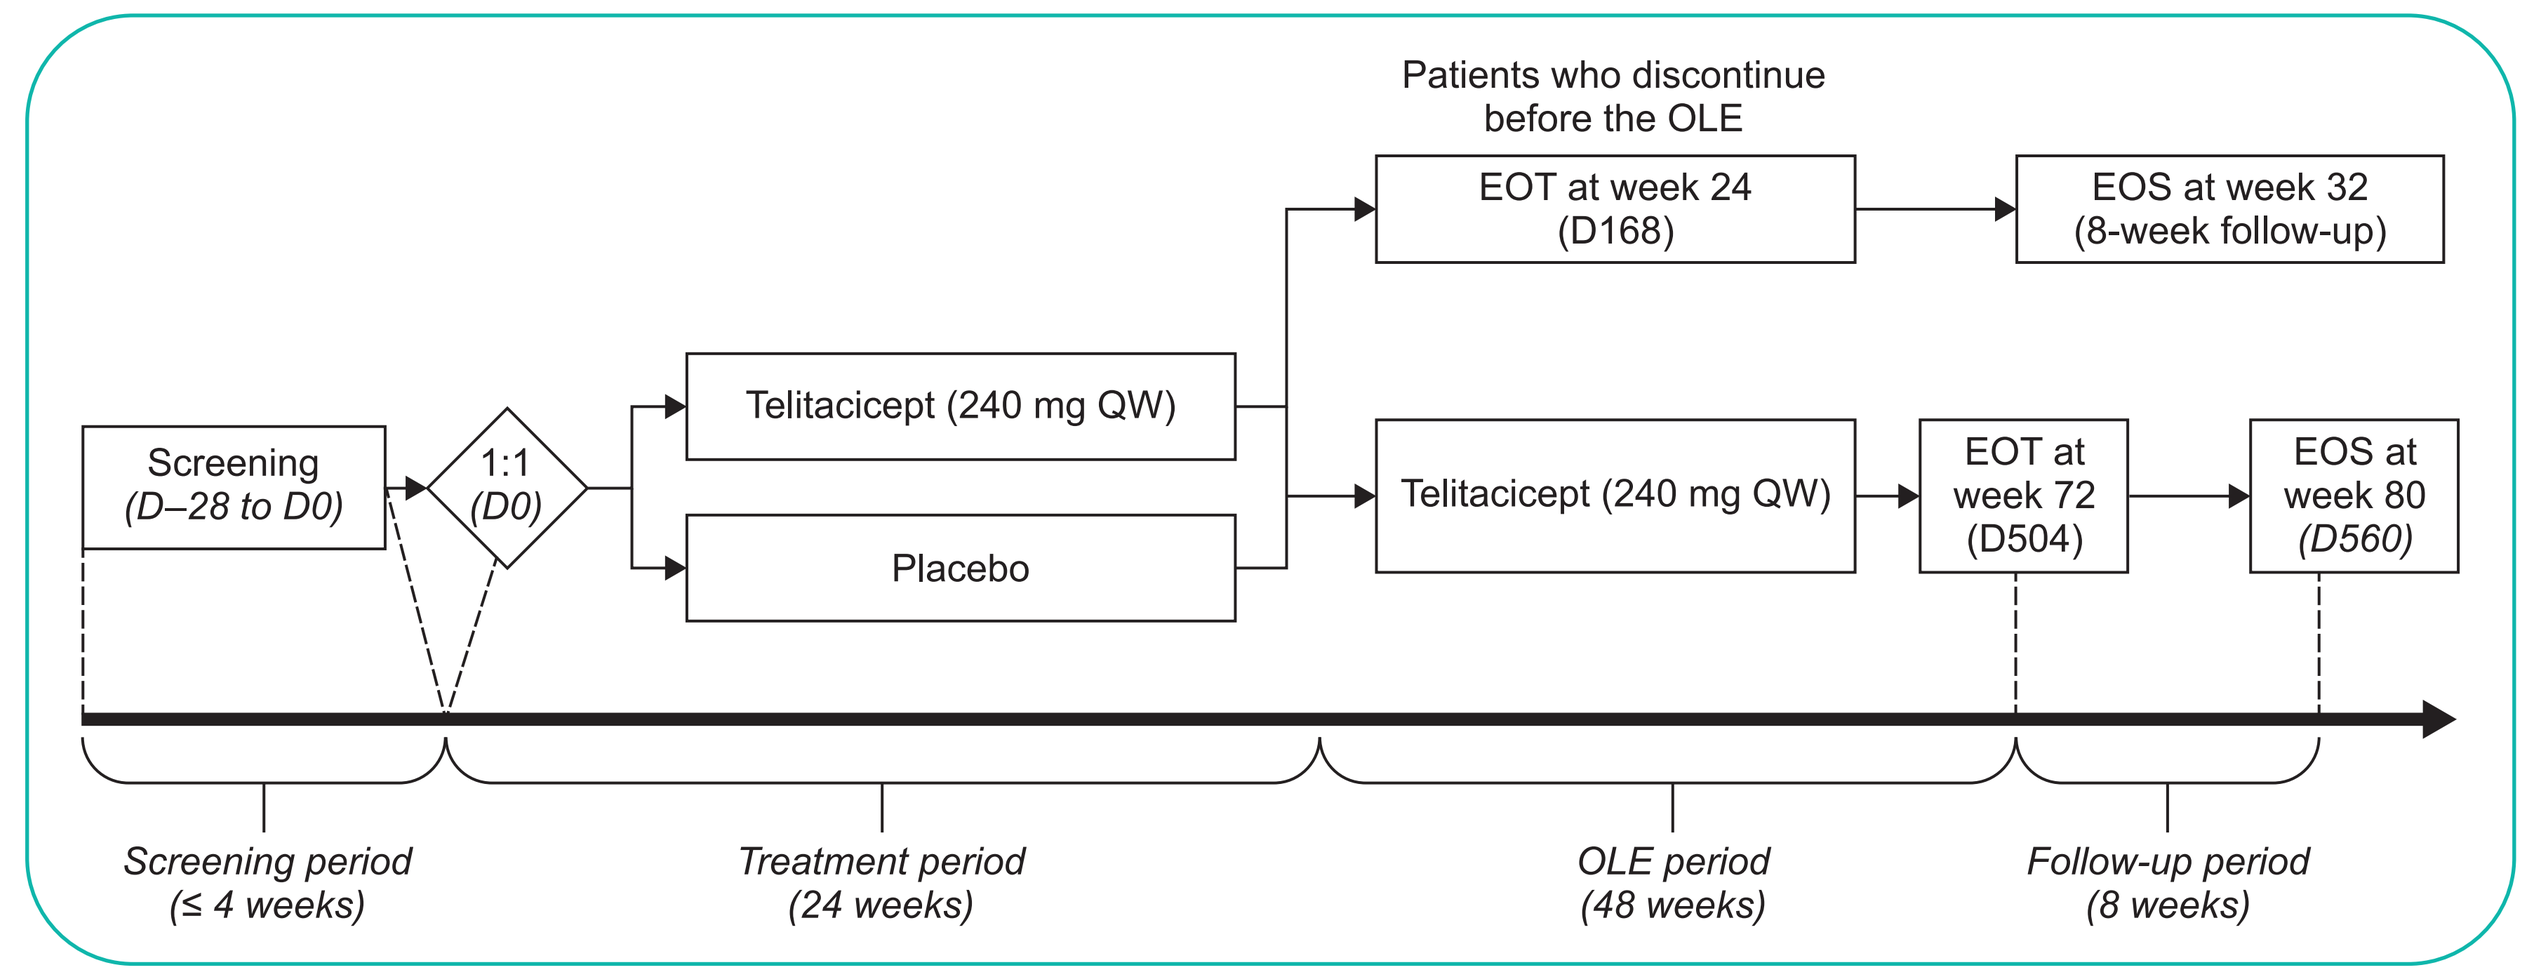
Patients who discontinue before the OLE
EOT at week 24 (D168)
EOS at week 32 (8-week follow-up)
Screening (D–28 to D0)
1:1 (D0)
Telitacicept (240 mg QW)
Placebo
Telitacicept (240 mg QW)
EOT at week 72 (D504)
EOS at week 80 (D560)
Screening period (≤ 4 weeks)
Treatment period (24 weeks)
OLE period (48 weeks)
Follow-up period (8 weeks)

Figure 2. Study design. D=Day; **EOS**=end of study; **EOT**=end of treatment; **OLE**=open-label extension; **QW**=once weekly.

- **Patient population:** Adults with gMG diagnosis, on stable standard treatment, with positive serum AChR or MuSK antibody, Myasthenia Gravis Foundation of America (MGFA) class II, III, or IVa, Myasthenia Gravis-Activities of Daily Living (MG-ADL) score ≥6, and Quantitative Myasthenia Gravis (QMG) score ≥8.
- **Assessments:** The primary endpoint was change in MG-ADL at week 24. Secondary endpoints included change in QMG at week 24, the proportion of patients with ≥3-point reduction in MG-ADL, and the proportion of patients with ≥5-point reduction in QMG. Safety and immunological parameters were assessed.

Results

- 114 patients (demographics in **Table 1**) were randomized (telitacicept, n=57; placebo, n=57); 54 patients in the telitacicept arm and 52 patients in the placebo arm completed the study (**Figure 3**).

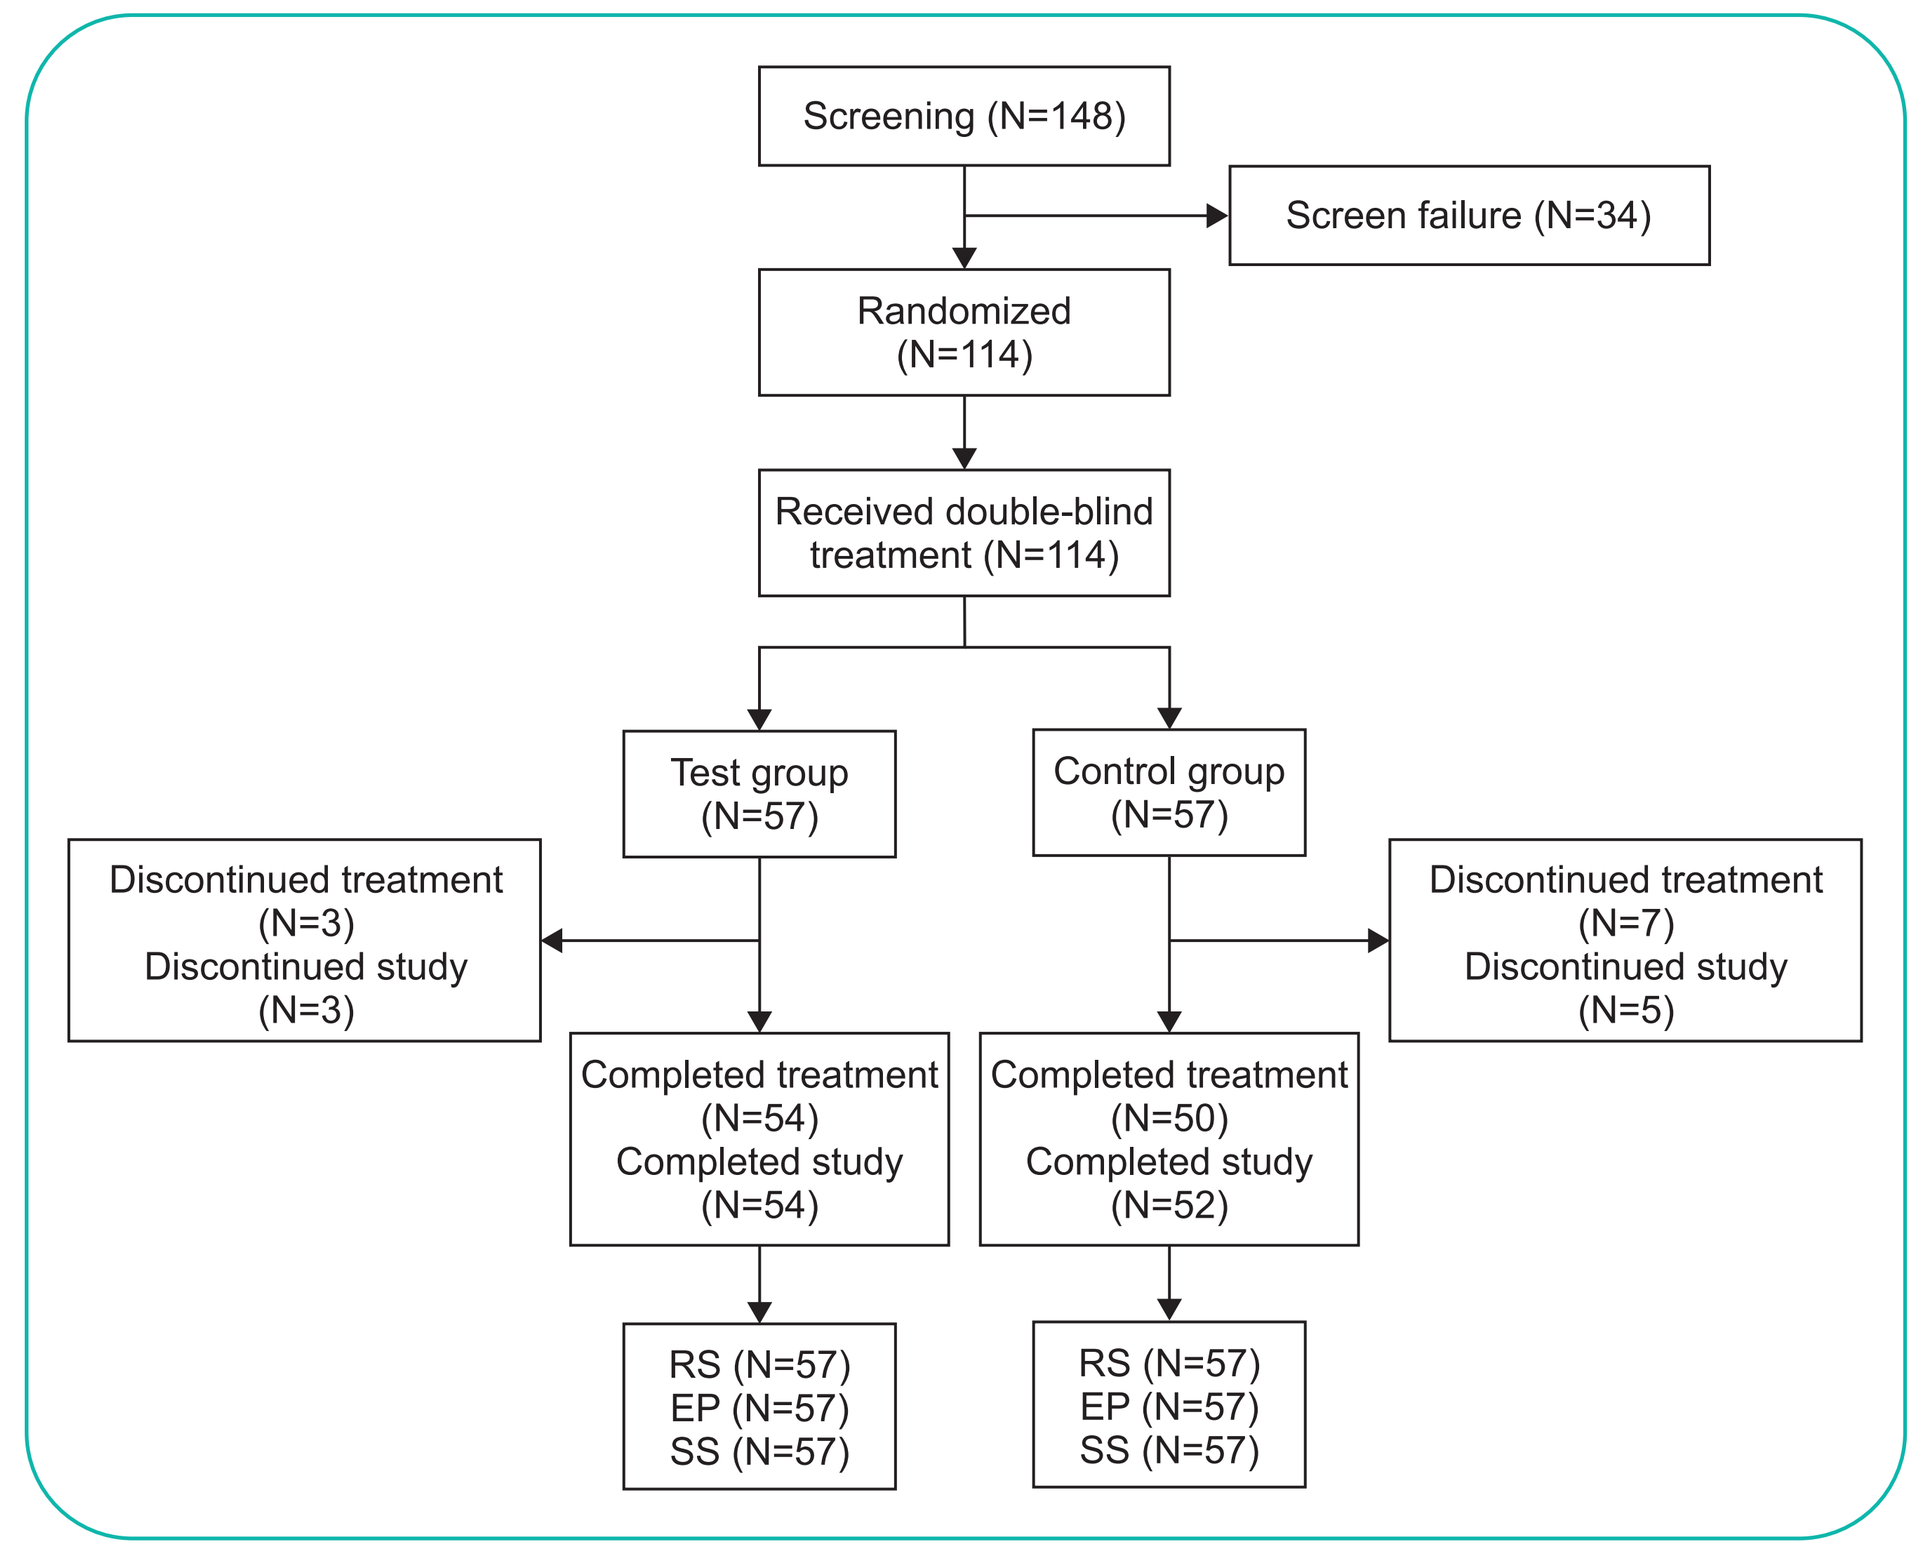
Screening (N=148)
Screen failure (N=34)
Randomized (N=114)
Received double-blind treatment (N=114)
Test group (N=57)
Control group (N=57)
Discontinued treatment (N=3) Discontinued study (N=3)
Discontinued treatment (N=7) Discontinued study (N=5)
Completed treatment (N=54) Completed study (N=54)
Completed treatment (N=50) Completed study (N=52)
RS (N=57) EP (N=57) SS (N=57)
RS (N=57) EP (N=57) SS (N=57)

Figure 3. Patient disposition. EP=efficacy population; **RS**=randomized set; **SS**=safety set.

	Telitacicept (N=57)	Placebo (N=57)
Age (years), mean ± SD	49.1 ± 14.69	49.6 ± 15.03
Sex		
Male, n (%)	30 (52.6)	21 (36.8)
Female, n (%)	27 (47.4)	36 (63.2)
Disease duration (month), mean ± SD	83.09 ± 84.507	76.05 ± 87.817
MGFA classification		
Class IIa, n (%)	3 (5.3)	12 (21.1)
Class IIb, n (%)	14 (24.6)	9 (15.8)
Class IIIa, n (%)	25 (43.9)	23 (40.4)
Class IIIb, n (%)	11 (19.3)	12 (21.1)
Class IVa, n (%)	4 (7.0)	1 (1.8)
Baseline MG-ADL score, mean ± SD	10.0 ± 2.60	9.9 ± 2.62
Baseline QMG score, mean ± SD	17.9 ± 3.43	18.8 ± 3.65
Antibody-positive at screening		
AChR, n (%)	55 (96.5)	55 (96.5)
MuSK, n (%)	2 (3.6)	2 (3.5)
Standard-of-care therapy		
Anticholinesterase inhibitors, n (%)	53 (93.0)	52 (91.2)
Steroids, n (%)	36 (63.2)	34 (59.6)
Immunosuppressants, n (%)	34 (59.6)	34 (59.6)

Table 1. Patient demographics. AChR=acetylcholine receptor; **MG-ADL**=Myasthenia Gravis-Activities of Daily Living; **MGFA**=Myasthenia Gravis Foundation of America; **MuSK**=muscle-specific kinase; **QMG**=Quantitative Myasthenia Gravis; **SD**=standard deviation.

Primary endpoint

- At week 24, change in MG-ADL was –6.4 for telitacicept and –1.6 for placebo (p<0.001); with missing data imputed as non-response, change in MG-ADL was –5.74 and –0.91, respectively (p<0.001) (**Figure 4A**).

Secondary endpoints

- At week 24, change in QMG was –7.8 for telitacicept and –1.9 for placebo (p<0.001); with missing data imputed as non-response, change in QMG was –8.66 and –2.27, respectively (p<0.001) (**Figure 4B**). 98.1% of telitacicept and 12.0% of placebo patients had ≥3-point reduction in MG-ADL (**Figure 4C**), and 87.0% of telitacicept and 16.0% of placebo patients had ≥5-point reduction in QMG (**Figure 4D**).

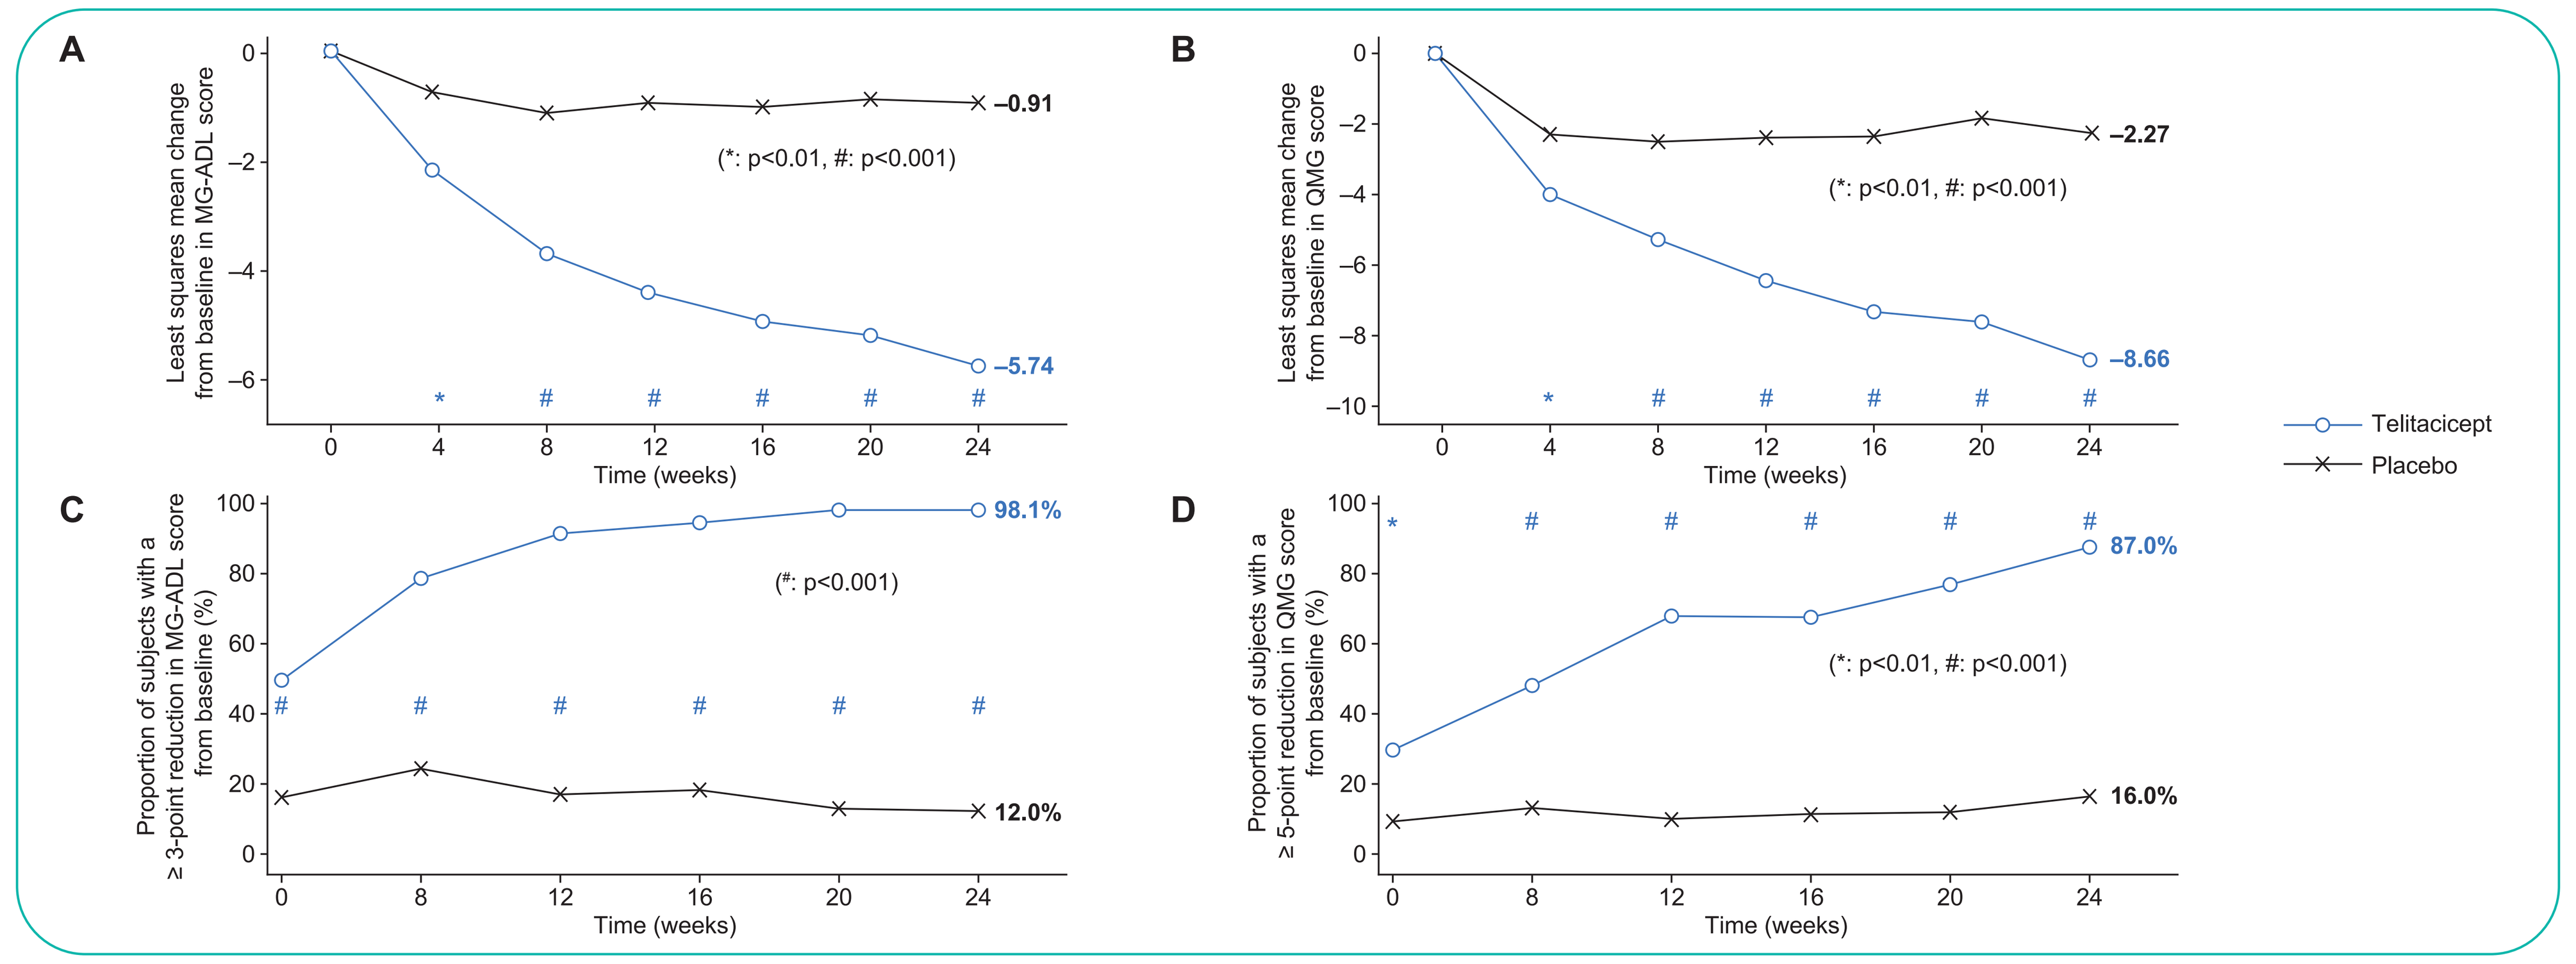
A
Least squares mean change from baseline in MG-ADL score
(*: p<0.01, #: p<0.001)
–0.91
–5.74
Time (weeks)
B
Least squares mean change from baseline in QMG score
(*: p<0.01, #: p<0.001)
–2.27
–8.66
Time (weeks)
Telitacicept
Placebo
C
Proportion of subjects with a ≥ 3-point reduction in MG-ADL score from baseline (%)
(#: p<0.001)
98.1%
12.0%
Time (weeks)
D
Proportion of subjects with a ≥ 5-point reduction in QMG score from baseline (%)
(*: p<0.01, #: p<0.001)
87.0%
16.0%
Time (weeks)

Figure 4. Change in MG-ADL from baseline (A), change in QMG from baseline (B), proportion of patients with ≥3-point reduction in MG-ADL from baseline (C), proportion of patients with ≥5-point reduction in QMG from baseline (D), (FAS). FAS=full analysis set; **MG-ADL**=Myasthenia Gravis-Activities of Daily Living; **QMG**=Quantitative Myasthenia Gravis.

- The proportion of patients with a more stringent level of improvements in MG-ADL or QMG score at week 24 is shown in **Figure 5**.

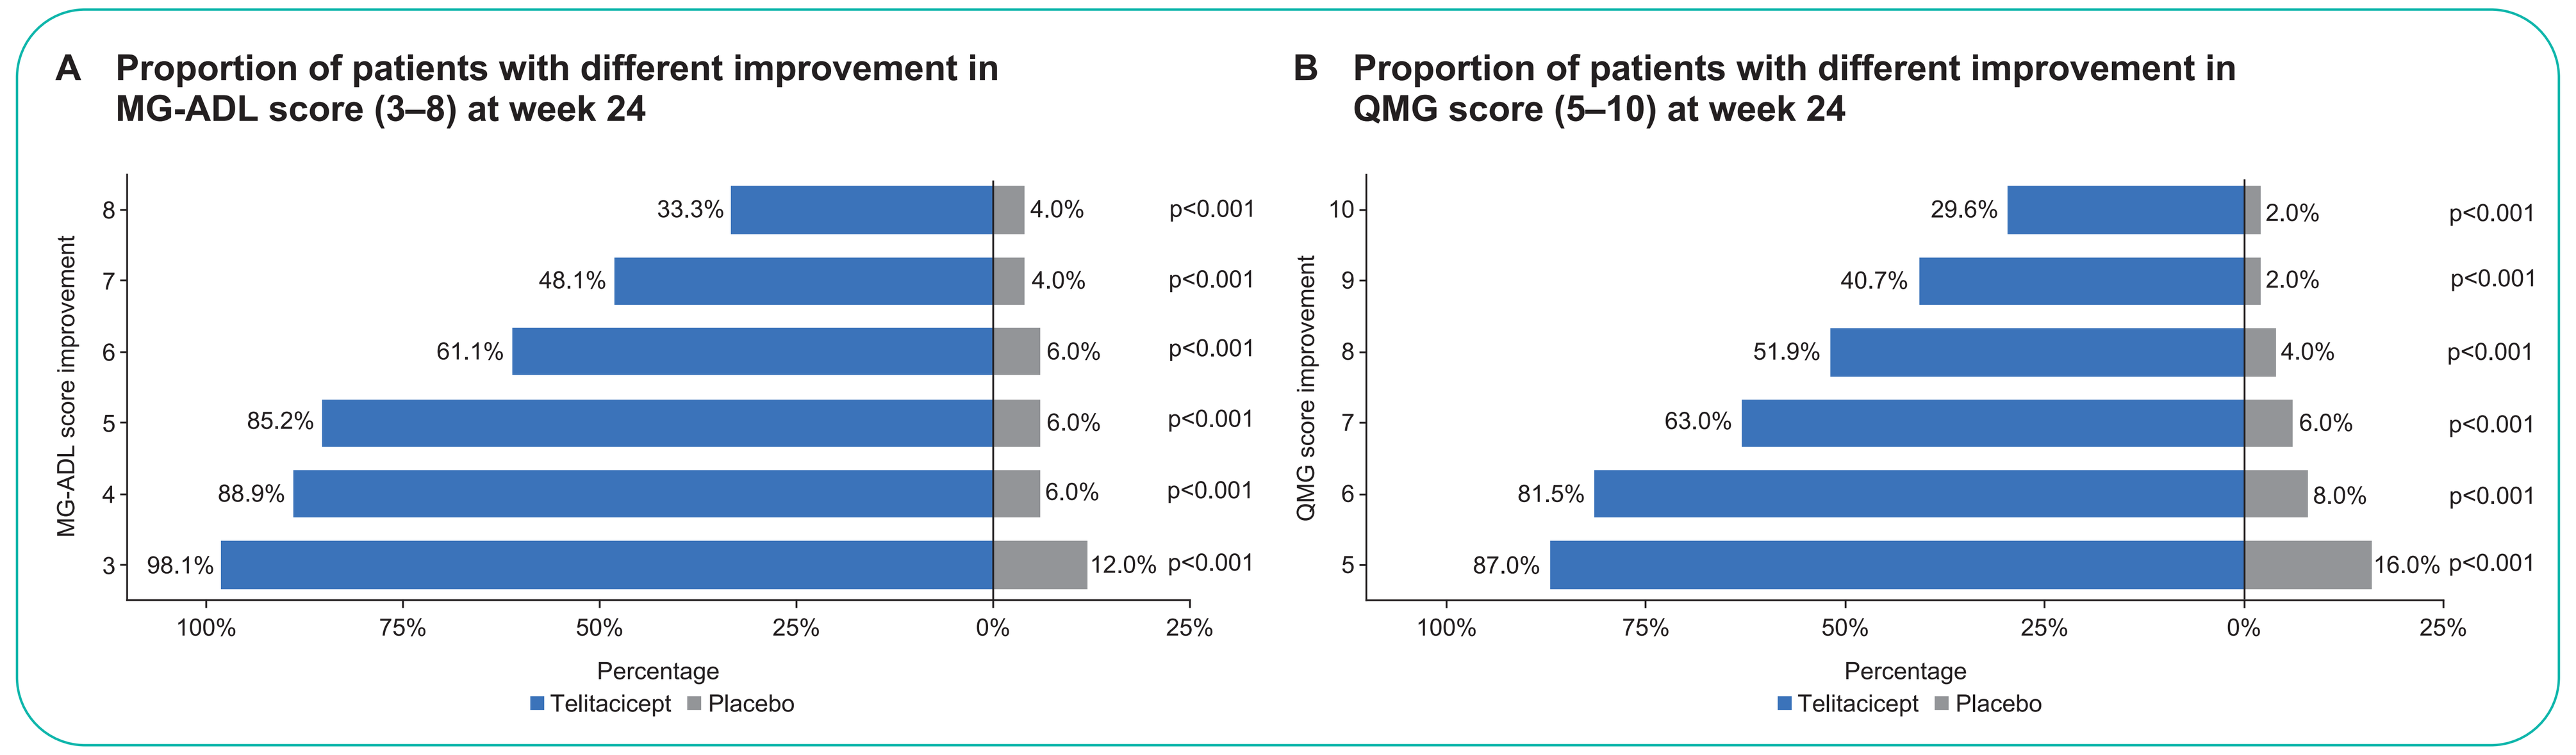
A Proportion of patients with different improvement in MG-ADL score (3–8) at week 24
MG-ADL score improvement
8: 33.3% / 4.0% p<0.001
7: 48.1% / 4.0% p<0.001
6: 61.1% / 6.0% p<0.001
5: 85.2% / 6.0% p<0.001
4: 88.9% / 6.0% p<0.001
3: 98.1% / 12.0% p<0.001
Percentage
Telitacicept Placebo
B Proportion of patients with different improvement in QMG score (5–10) at week 24
QMG score improvement
10: 29.6% / 2.0% p<0.001
9: 40.7% / 2.0% p<0.001
8: 51.9% / 4.0% p<0.001
7: 63.0% / 6.0% p<0.001
6: 81.5% / 8.0% p<0.001
5: 87.0% / 16.0% p<0.001
Percentage
Telitacicept Placebo

Figure 5. Proportion of patients with different levels of improvement at week 24 of MG-ADL score (A) and QMG score (B), (FAS). FAS=full analysis set; **MG-ADL**=Myasthenia Gravis-Activities of Daily Living; **QMG**=Quantitative Myasthenia Gravis.

Safety and immunological parameters

- The most frequently reported adverse event (AE) with telitacicept was IgM decrease (26.3%).
- The incidence of treatment-emergent AEs was similar between the telitacicept group (86.0%) and placebo group (82.5%).
- Infection-associated AEs are shown in **Table 2**; upper respiratory tract infection was the most common infectious AE reported in both treatment groups.
- Serious AEs are shown in **Table 3**.
- Injection site reactions were reported in 14.0% of the telitacicept group and 1.8% of the placebo group.

	Telitacicept (N=57)		Placebo (N=57)	
	n (%)	Events	n (%)	Events
Infections and infestations	26 (45.6)	46	34 (59.6)	50
Upper respiratory tract infection	12 (21.1)	17	20 (35.1)	24
Urinary tract infection	9 (15.8)	11	6 (10.5)	6
Pneumonia	1 (1.8)	1	6 (10.5)	6
Respiratory tract infection	1 (1.8)	1	2 (3.5)	2
Influenza	0 (0)	0	3 (5.3)	3

Table 2. Infection-associated AEs occurring in >5% of subjects (SS). AE=adverse event; **n**=number of subjects; **SS**=safety set.

	Telitacicept (N=57)		Placebo (N=57)	
	n (%)	Events	n (%)	Events
Serious AEs	4 (7.0)	4	6 (10.5)	7
Pneumonia	1 (1.8)	1	4 (7.0)	4
COVID-19 pneumonia	1 (1.8)	1	0 (0)	0
Influenza	0 (0)	0	1 (1.8)	1
Upper respiratory tract infection	0 (0)	0	1 (1.8)	1
Open fracture	0 (0)	0	1 (1.8)	1
Pneumonitis	1 (1.8)	1	0 (0)	0
Accidental death	1 (1.8)	1	0 (0)	0

Table 3. Serious AEs (SS). AE=adverse event; **n**=number of subjects; **SS**=safety set.

Conclusion

Telitacicept demonstrated statistically significant and clinically meaningful efficacy, and a safety profile consistent with data from clinical trials in systemic lupus erythematosus, rheumatoid arthritis, progressive systemic sclerosis and IgA neuropathy, and post-marketing data.

A global phase 3 pivotal trial (RemeMG) is evaluating the efficacy and safety of telitacicept in gMG in a diverse global patient population. The RemeMG study is currently ongoing and recruiting patients.

Presented at the American Academy of Neurology (AAN) Annual Meeting 2025, San Diego, USA, April 5–9, 2025

References: 1. Yin J, *et al. Eur J Neurol.* 2024;31(8):e16322. **Disclosures:** **JY** is a consultant in a global scientific advisor committee for Alexion. **WW**, **LL**, **YS**, and **JF** are employees of RemeGen Co., Ltd, and receive company stock. **QZ** is an employee of RemeGen Biosciences and receives company stock. **Presenter Contact:** Jian Yin m13611177824@163.com