Telitacicept as a Novel BAFF/APRIL-Targeted Therapy for Sjögren's Disease

Sambhawana Bhandari,[1] Himanshu Patel,[2] Jeremy Sokolove,[2] Alfred HJ Kim[1]

[1]Division of Rheumatology, Department of Medicine, Washington University School of Medicine, St. Louis, MO, USA; [2]Vor Biopharma Inc., Boston, MA, USA

BACKGROUND

- Sjögren disease (SjD) is a systemic autoimmune disease in which autoreactive B cells target and damage the epithelial cells of exocrine glands[1-4]
- Lymphocytic infiltration of exocrine glands such as salivary and lachrymal glands results in the predominant characterizations of xerostomia, keratoconjunctivitis sicca, and fatigue; approximately 30% of patients develop additional systemic manifestations[1-5]
- Pathologic B-cell and plasma cell activation contributes to SjD pathogenesis and drives tissue injury by promoting autoantibody production, immune complex formation, and antibody-independent mechanisms such as T-cell activation and cytokine production, ultimately leading to disease progression, systemic complications, and increased risk of lymphoma[1-3]
- Current therapies for SjD largely remain symptomatic or are associated with non-selective immunosuppression, representing a high unmet need for innovative and effective treatment options[1,4]
- Telitacicept is a novel, fully human, TACI-Fc fusion protein that targets B-cell activating factor (BAFF) and a proliferation-inducing ligand (APRIL)[4,6,7]
- Through this novel dual inhibition mechanism, telitacicept modulates upstream B-cell development and survival, which ultimately may reduce pathogenic autoantibodies implicated in autoimmune disease, including SjD (**Figure 1**)[4,6,7]

Figure 1. Telitacicept Is a Fusion Protein Based on the Human TACI Receptor Designed to Target BAFF and APRIL

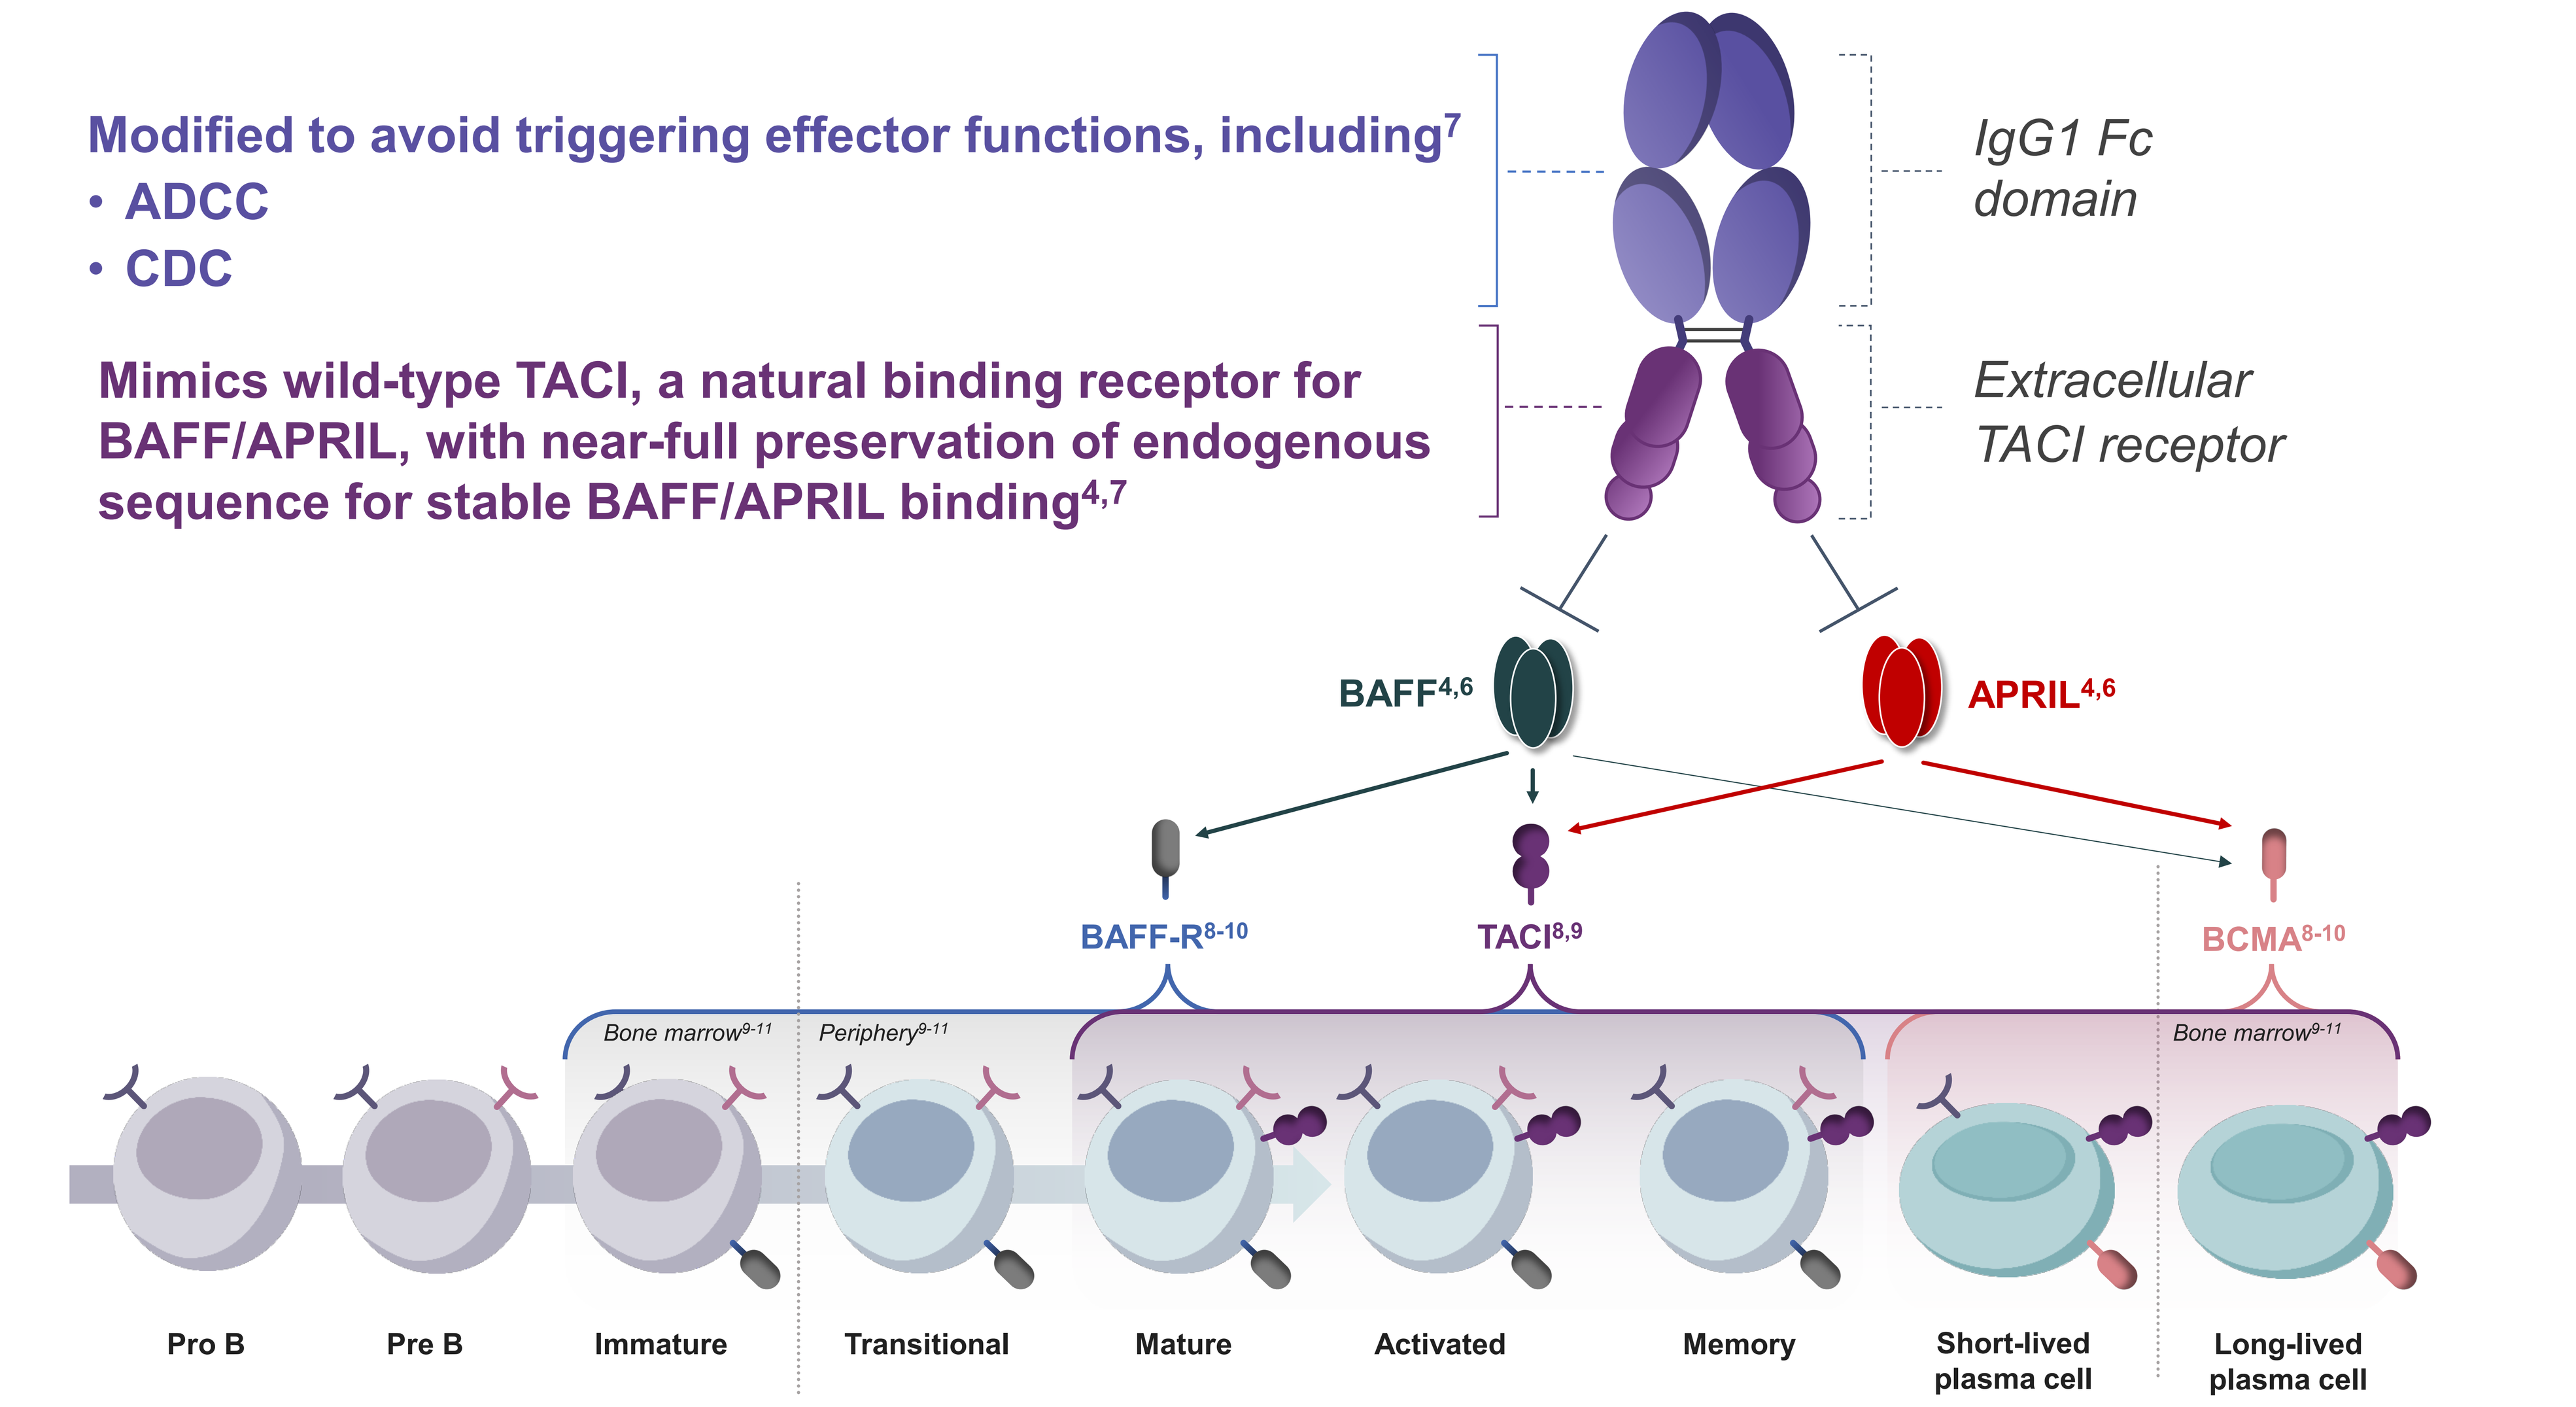
Modified to avoid triggering effector functions, including[7]
• ADCC
• CDC
Mimics wild-type TACI, a natural binding receptor for BAFF/APRIL, with near-full preservation of endogenous sequence for stable BAFF/APRIL binding[4,7]
IgG1 Fc domain
Extracellular TACI receptor
BAFF[4,6]
APRIL[4,6]
BAFF-R[8-10]
TACI[8,9]
BCMA[8-10]
Bone marrow[9-11]
Periphery[9-11]
Bone marrow[9-11]
Pro B
Pre B
Immature
Transitional
Mature
Activated
Memory
Short-lived plasma cell
Long-lived plasma cell

ADCC, antibody-dependent cellular cytotoxicity; APRIL, a proliferation-inducing ligand; BAFF, B-cell activating factor; BAFF-R, B-cell activating factor receptor; BCMA, B-cell maturation antigen; CDC, complement-dependent cytotoxicity; Fc, fragment crystallizable; IgG1, immunoglobulin G1; TACI, transmembrane activator and calcium-modulator and cyclophilin ligand (CAML) interactor.

- Data from phase 2 (NCT04078386) and phase 3 (NCT05673993) studies of telitacicept conducted in China and sponsored by RemeGen Co., Ltd. showed efficacy and safety in adults with primary SjD[12-15]
- Here, we present the pharmacodynamic findings from a phase 2 study of telitacicept in adults with primary SjD conducted in China

METHODS

Study Design

- Eligible participants were randomly assigned (1:1:1) to receive subcutaneous telitacicept (240 mg), subcutaneous telitacicept (160 mg), or placebo for 24 weeks, once weekly (**Figure 2**)
- Eligibility criteria included participants with primary SjD who fulfilled the 2016 American College of Rheumatology (ACR)/European Alliance of Associations for Rheumatology (EULAR) classification criteria for SjD, were aged 18-65 years, had anti-Sjögren's syndrome–related antigen A (SSA) antibody positivity, and a EULAR Sjögren's Syndrome Disease Activity Index (ESSDAI) score of ≥5 points
- Participants were not permitted to receive concomitant treatment for SjD except hydroxychloroquine if stable for >12 weeks before randomization

Figure 2. Study Design

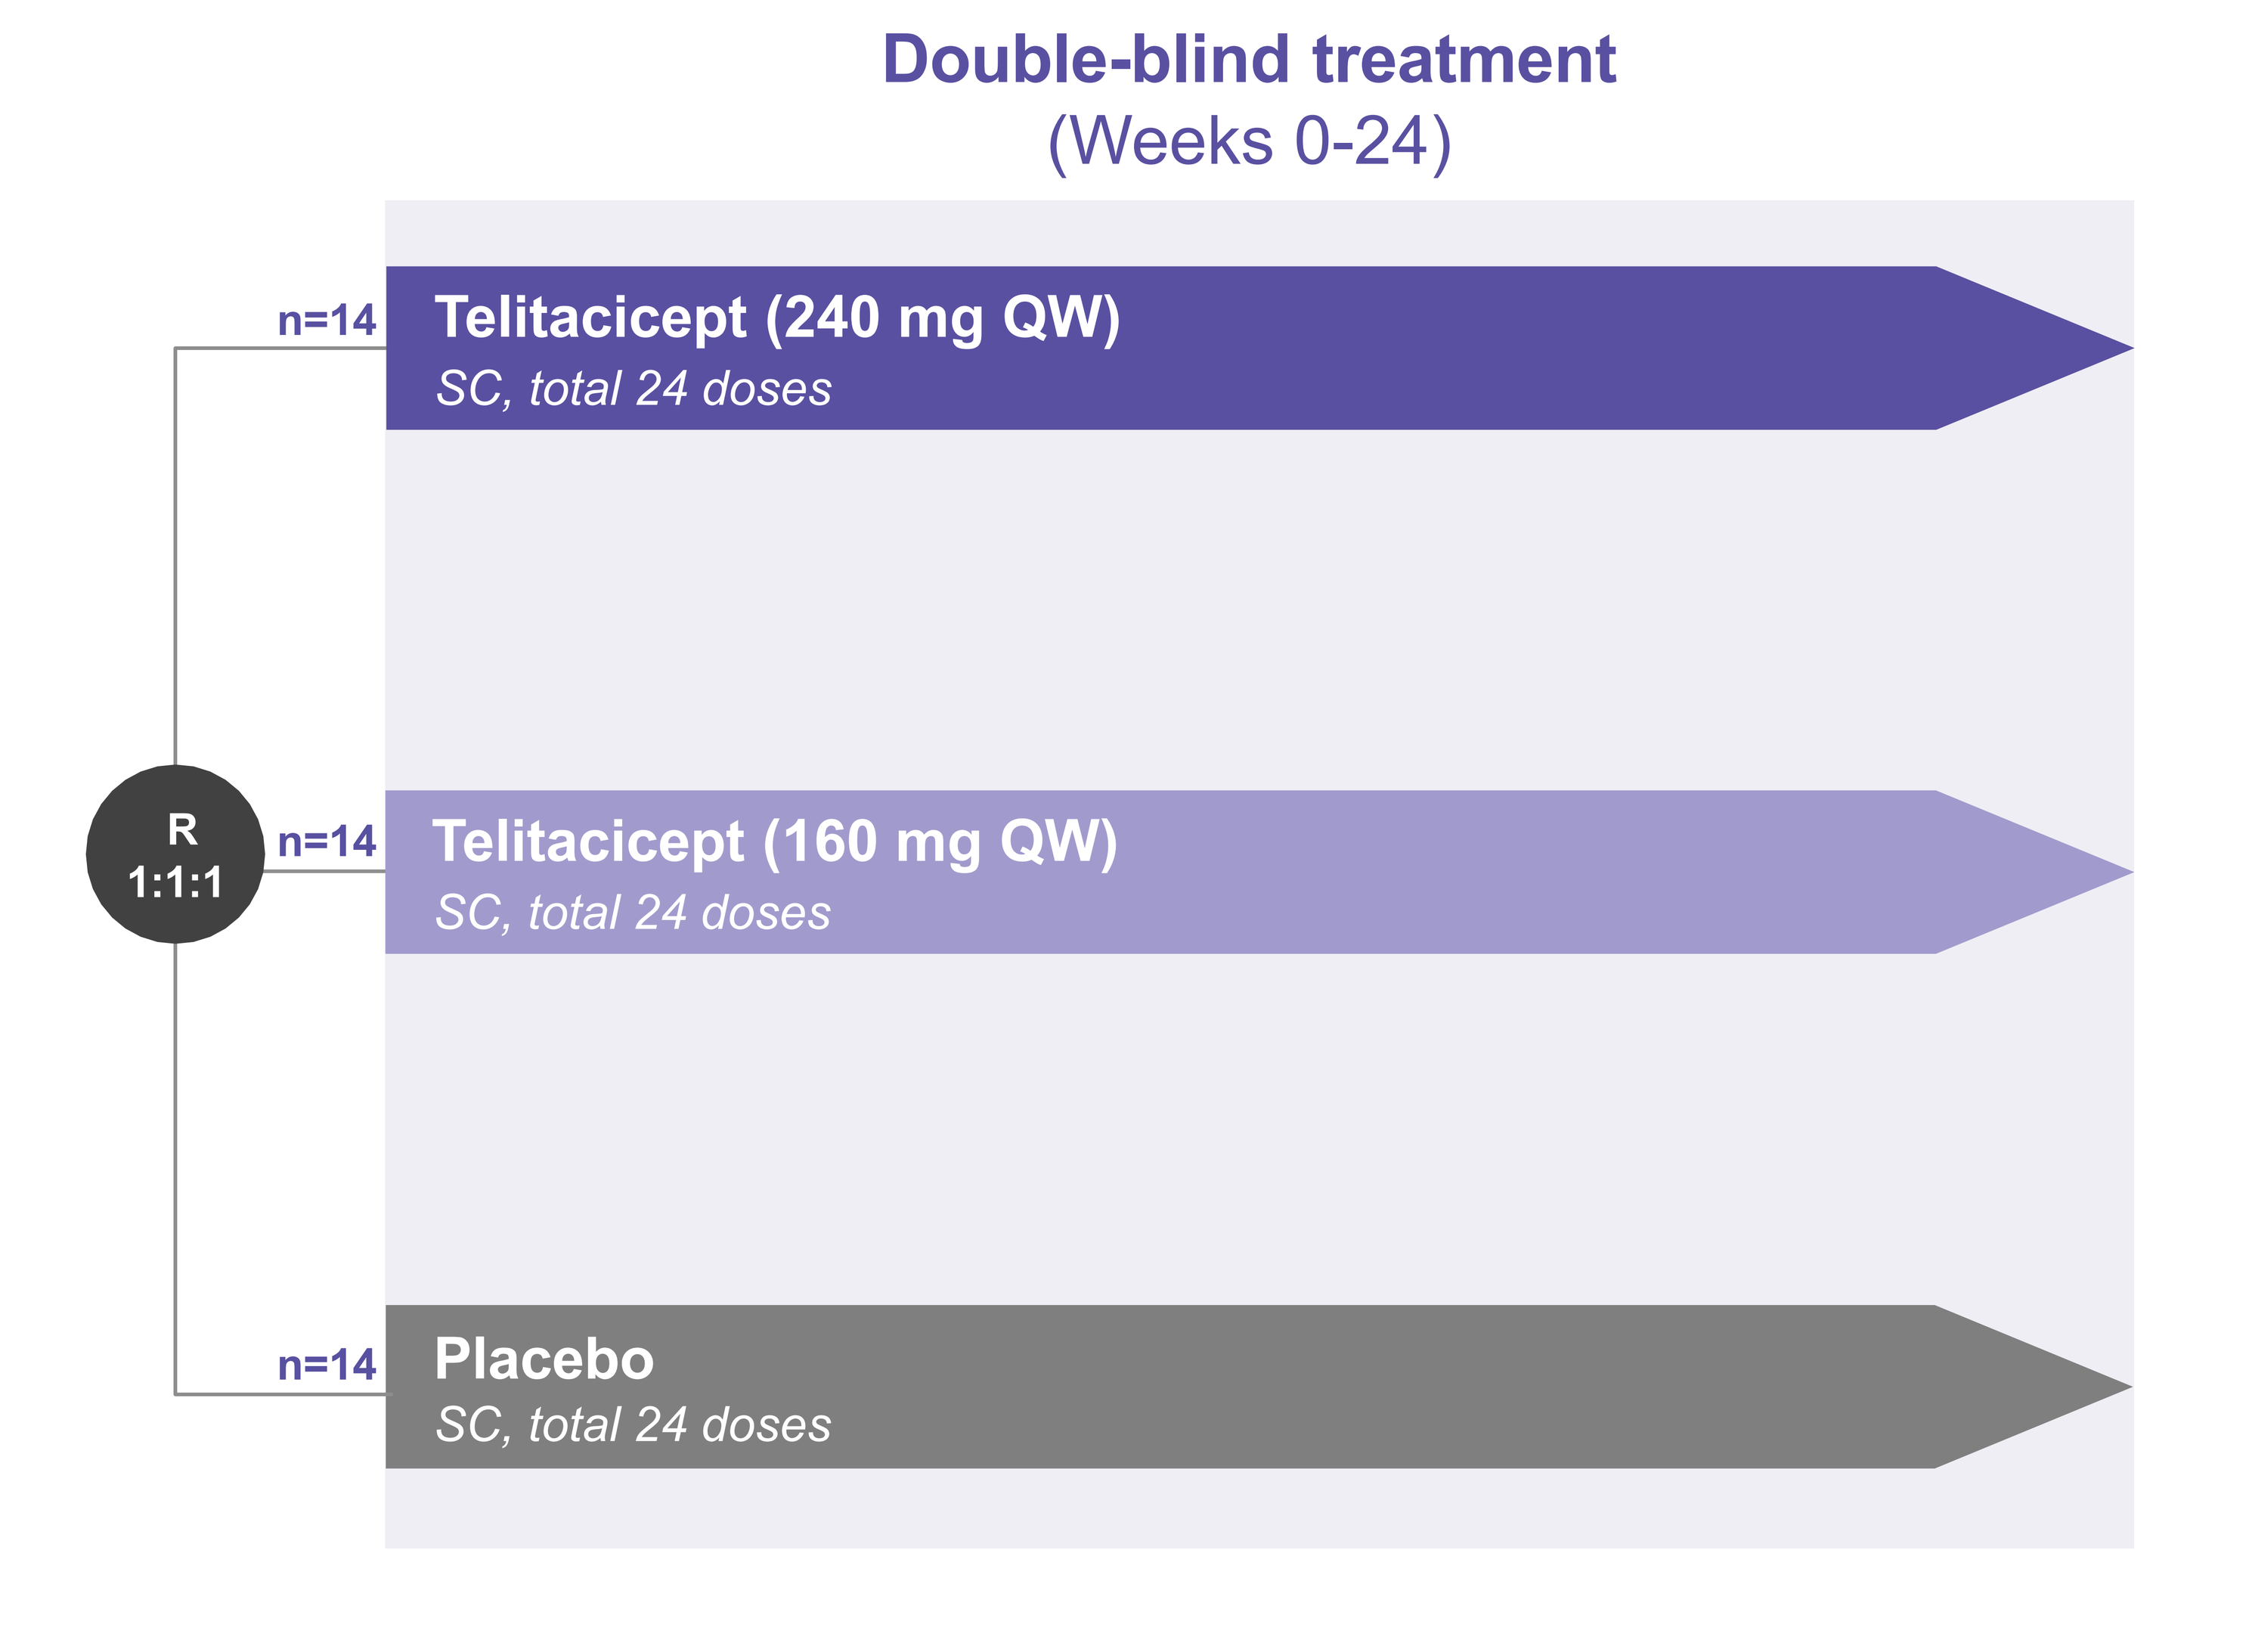
Double-blind treatment
(Weeks 0-24)
R
1:1:1
n=14
Telitacicept (240 mg QW)
SC, total 24 doses
n=14
Telitacicept (160 mg QW)
SC, total 24 doses
n=14
Placebo
SC, total 24 doses

QW, once weekly; R, randomization; SC, subcutaneous.

Endpoints

- The primary endpoint was the change from baseline in the ESSDAI score at week 24
- Biomarkers included immunoglobulin G (IgG), IgA, IgM, C3, C4, CD19+ B cells, CD4+ T cells, and CD8+ T cells
- Safety assessments included the incidence and severity of adverse events (AEs)

RESULTS

Efficacy

- Telitacicept was associated with reductions in serum IgG, IgA, and IgM, and CD19+ B-cell counts compared with placebo (**Figure 3**)
- At week 24, IgG levels reduced from 19.6 to 15.6 g/L, IgA from 3.0 to 1.5 g/L, and IgM from 1.2 to 0.5 g/L, while circulating CD19+ B-cell counts decreased from 157.8 to 81.7 cells/µL in participants treated with telitacicept 160 mg (**Table 1**)
- The primary endpoint of change from baseline in ESSDAI score at week 24 was met with statistical significance, and the efficacy of telitacicept was replicated across multiple endpoints in a phase 3 randomized controlled study conducted in China with 380 participants with similar changes in pharmacodynamics

Figure 3. Pharmacodynamic Parameters: Change From Baseline in Immunoglobulins and CD19+ B Cells

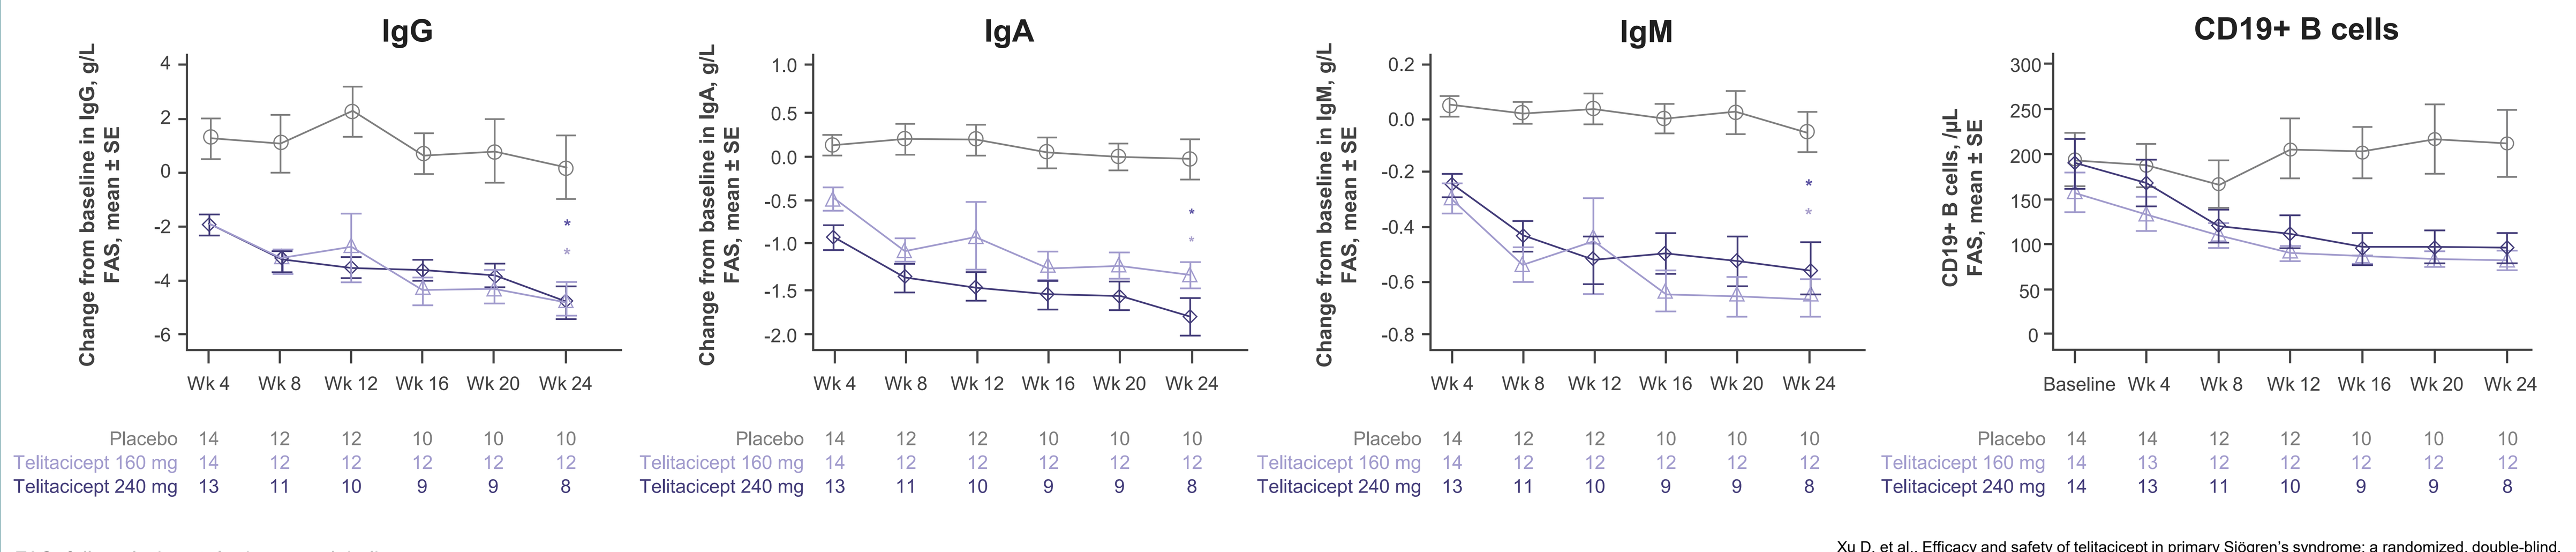
IgG
Change from baseline in IgG, g/L FAS, mean ± SE
IgA
Change from baseline in IgA, g/L FAS, mean ± SE
IgM
Change from baseline in IgM, g/L FAS, mean ± SE
CD19+ B cells
CD19+ B cells, /µL FAS, mean ± SE
Wk 4 Wk 8 Wk 12 Wk 16 Wk 20 Wk 24
Baseline Wk 4 Wk 8 Wk 12 Wk 16 Wk 20 Wk 24
Placebo 14 12 12 10 10 10
Telitacicept 160 mg 14 12 12 12 12 12
Telitacicept 240 mg 13 11 10 9 9 8
Placebo 14 14 12 12 10 10 10
Telitacicept 160 mg 14 13 12 12 12 12 12
Telitacicept 240 mg 14 13 11 10 9 9 8
Placebo (n=14)
Telitacicept 160 mg (n=14)
Telitacicept 240 mg (n=14)

FAS, full analysis set; Ig, immunoglobulin.
*P<0.001 vs placebo.

Xu D, et al., Efficacy and safety of telitacicept in primary Sjögren's syndrome: a randomized, double-blind, placebo-controlled, phase 2 trial, *Rheumatology (Oxford)*, 2024;63(3):698-705, Copyright © 2023, © Xu D, et al. 2023. by permission of Oxford University Press on behalf of British Society for Rheumatology.

Table 1. Mean Change in Immunoglobulins and CD19+ B-Cell Counts at Week 24 Compared With Baseline

Mean (SD)	Placebo		Telitacicept 160 mg	
	Baseline	Week 24	Baseline	Week 24
IgG, g/L	16.3 (4.6)	16.9 (6.3)	19.6 (6.1)	15.6 (5.4)*
IgA, g/L	3.5 (1.6)	3.7 (1.7)	3.0 (1.2)	1.5 (0.6)*
IgM, g/L	1.3 (0.7)	1.0 (0.7)	1.2 (0.4)	0.5 (0.3)*
CD19+ B cells, /µL	193.7 (112.5)	211.6 (118.0)	157.8 (83.2)	81.7 (35.3)

Ig, immunoglobulin.
*Statistically significant compared with placebo (*P*<0.001).

Safety

- Telitacicept showed a generally well-tolerated safety profile in participants with primary SjD, with most AEs being mild or moderate in severity (**Table 2**)
- No deaths or serious AEs occurred in the telitacicept treatment groups

Table 2. Selected Adverse Events That Occurred in ≥2 Participants in Any Group

System organ class preferred term, n (%)	Placebo (n=14)	Telitacicept 160 mg (n=14)
Adverse events	13 (92.9)	12 (85.7)
Adverse reactions	9 (64.3)	9 (64.3)
Infections and infestations	8 (57.1)	9 (64.3)
General disorders and administration site conditions	0 (0)	7 (50.0)[a,*]

CTCAE, Common Terminology Criteria for Adverse Events.
[a]Included 6 instances of injection site reactions (CTCAE grade 1-2).
*Statistically significant compared with placebo (*P*<0.05).

CONCLUSIONS

- BAFF/APRIL inhibition may provide meaningful clinical benefit in SjD while largely preserving overall B-cell and Ig compartments, consistent with selective modulation of pathogenic B-cell populations
- A global, multicenter, randomized, double-blind, placebo-controlled phase 3 study evaluating the efficacy and safety of telitacicept in adult participants with active primary SjD is currently recruiting (NCT07404865; UPSTREAM SjD)
- Results from a global adult primary SjD population will further characterize the efficacy and safety of telitacicept

VOR

Presented at CCR-East 2026; April 30-May 3, 2026; Destin, FL

Disclosures: **Sambhawana Bhandari** has no disclosures. **Himanshu Patel** and **Jeremy Sokolove** are employees of Vor Biopharma Inc. and own company stock. **Alfred HJ Kim** receives grant support from AstraZeneca, Bristol Myers Squibb, CRISPR Therapeutics, and Novartis. AHJK also receives consulting fees from AbbVie, AstraZeneca, Aurinia Pharmaceuticals, Biogen, Exagen Diagnostics, Genentech/Roche, GlaxoSmithKline, Hinge Bio, Invivyd, Johnson & Johnson, Kymera Therapeutics, Kypha, Miltenyi Biomedicine, Novartis, and Zenas Biopharma. AHJK is a patent beneficiary with Kypha (US patent 10029318B2).

References: 1. Baldini C, et al. *Clin Exp Rheumatol*. 2024;42(12):2336-2343. 2. Maslinska M, Kostyra-Grabczak K. *Front Immunol*. 2024;15:1376723. 3. Verstappen GM, et al. *Nat Rev Rheumatol*. 2021;17(6):333-348. 4. Zeng L, et al. *J Autoimmun*. 2024;148:103291. 5. Seror R, et al. *Arthritis Care Res (Hoboken)*. 2013;65(8):1358-1364. 6. Ji L, et al. *MedComm (2020)*. 2024;5(4):e515. 7. European Patent Application EP3868403A1. 8. Tagami N, et al. *Int J Clin Oncol*. 2024;29(11):1676-1683. 9. Smulski CR, Eibel H. *Front Immunol*. 2018;9:2285. 10. Abeles I, et al. *Annu Rev Immunol*. 2024;42(1):103-126. 11. Vincent FB, et al. *Nat Rev Rheumatol*. 2014;10(6):365-373. 12. ClinicalTrials.gov identifier: NCT04078386. Updated January 11, 2022. Accessed March 12, 2026. https://clinicaltrials.gov/study/NCT04078386. 13. ClinicalTrials.gov identifier: NCT05673993. Updated October 1, 2025. Accessed March 12, 2026. https://clinicaltrials.gov/study/NCT05673993. 14. Xu D, et al. Efficacy and safety of telitacicept in patients with Sjögren's disease: results from a multicenter, randomized, double-blind, placebo-controlled, phase 3 clinical study. Presented at: American College of Rheumatology Convergence 2025; October 24-29, 2025; Chicago, IL. 15. Xu D, et al. *Rheumatology (Oxford)*. 2024;63(3):698-705.

Acknowledgments: This study was funded by Vor Biopharma Inc., Boston, MA, USA. Editorial support was provided by Daniel Farkas, PhD, of Envision 90TEN, an Envision Medical Communications agency, a part of Envision Pharma Group, and was funded by Vor Biopharma Inc.